DUAL ANTIGEN TARGETING WITH CO-TRANSDUCED CD19/22 CAR T CELLS FOR RELAPSED/REFRACTORY ALL

Sara Ghorashian[1,2], Giovanna Lucchini[3], Rachel Richardson[4], Kyvi Nguyen[4], Craig Terris[4], Macarena Oporto-Espuelas[4], Jenny Yeung[4], Danielle Pinner[3], Jan Chu[3], Lindsey Williams[3], Ka-yuk Ko[3], Chloe Walding[8], Kelly Watts[9], Sarah Inglott[1], Stuart Adams [1], Emma Gravett[1], Kimberly Gilmour[10], Alka Lal[11], Sangeetha Kunaseelan[11], Bilyana Popova[11], Andre Lopes[11], Yenting Ngai[11], Evangelia K Kokalaki[12], Milena Balasch Carulla[3], Khushnuma Mullanfiroze[3], Arina Lazareva[3], Vesna Pavasovic[1], Anupama Rao[1], Jack Bartram[1], Ajay Vora[1], Robert Chiesa[3], Juliana Silva[3], Kanchan Rao[3], Katherine Clesham[8], Denise Bonney[9], Robert Wynn[9], Martin Pule[13], Rachael E Hough[8] and Persis J Amrolia[3]

1Department of Hematology, Great Ormond Street Hospital; 2 Developmental Biology and Cancer, UCL Great Ormond Street Institute of Child Health; 3 Bone Marrow Transplantation, Great Ormond Street Hospital for Children; 4 Molecular and Cellular Immunology Section, UCL Great Ormond Street Institute of Child Health; 8 Cancer Clinical Trials Unit, University College London Hospitals NHS Trust ;9 Bone Marrow Transplantation, Royal Manchester Children's Hospital; 10 Department of Immunology, Great Ormond Street Hospital for Children; 11 CRUK Cancer Trials Centre, University College London, London, United Kingdom; 12 Autolus Ltd, London, United Kingdom; 13 Cancer Institute, University College London

INTRODUCTION

We previously developed a novel fast off-rate CD19CAR (AUTO1) in R/R paediatric ALL (CARPALL study, NCT02443831). As a single-targeting CD19CAR T-cell therapy, we demonstrated excellent safety and CART expansion with persistence ongoing (up to 6 years)

CD19-ve relapse was the major cause of treatment failure (5/6 relapses, of 14 treated patients), as for other CD19CAR T-cell therapies including Kymriah

We therefore investigated dual targeting approaches. We developed a novel CD22CAR which shows potent responses even at low antigen levels (<500 copies/cell)

AIM

To investigate dual targeting of CD19/CD22 in a phase 1 CAR T cell study of R/R paediatric ALL

METHODS

GMP manufacture

A dual (mixed vector) lenti-transduction protocol was validated

Clinical study

We carried out a UK, multi-centre, non-randomised, open label Phase I clinical study conducted in 3 hospitals.

Eligible patients were children and young adults (age ≤24 years) with high risk, relapsed CD19+ and / or CD22+ B lineage ALL (the CARPALL study NCT02443831) and ineligible for Kymriah on the UK national access programme

CAR T persistence assays

These were performed on peripheral blood and bone marrow:

1) By flow cytometry of using 2 anti-idiotype antibodies, specific for each of the CARs
2) By qPCR, using probes specific to each of the transgenes

Analysis of CAR T cell kinetics

These were performed on the CAR transgene quantitation by qPCR. Area under the curve was estimated by a trapezoidal algorithm

Endpoints

Primary: NCI grade 3-5 toxicity, proportion in molecular CR at 1 month

Secondary: Proportion in molecular CR at 2 yrs without further therapy, relapse rate, antigen –ve relapse rate, EFS and OS at 1 and 2 yrs

Events were defined as per ELIANA (non-response, relapse, death). Stringent EFS also included emergence of MRD and further therapy as events

TRIAL SCHEMA

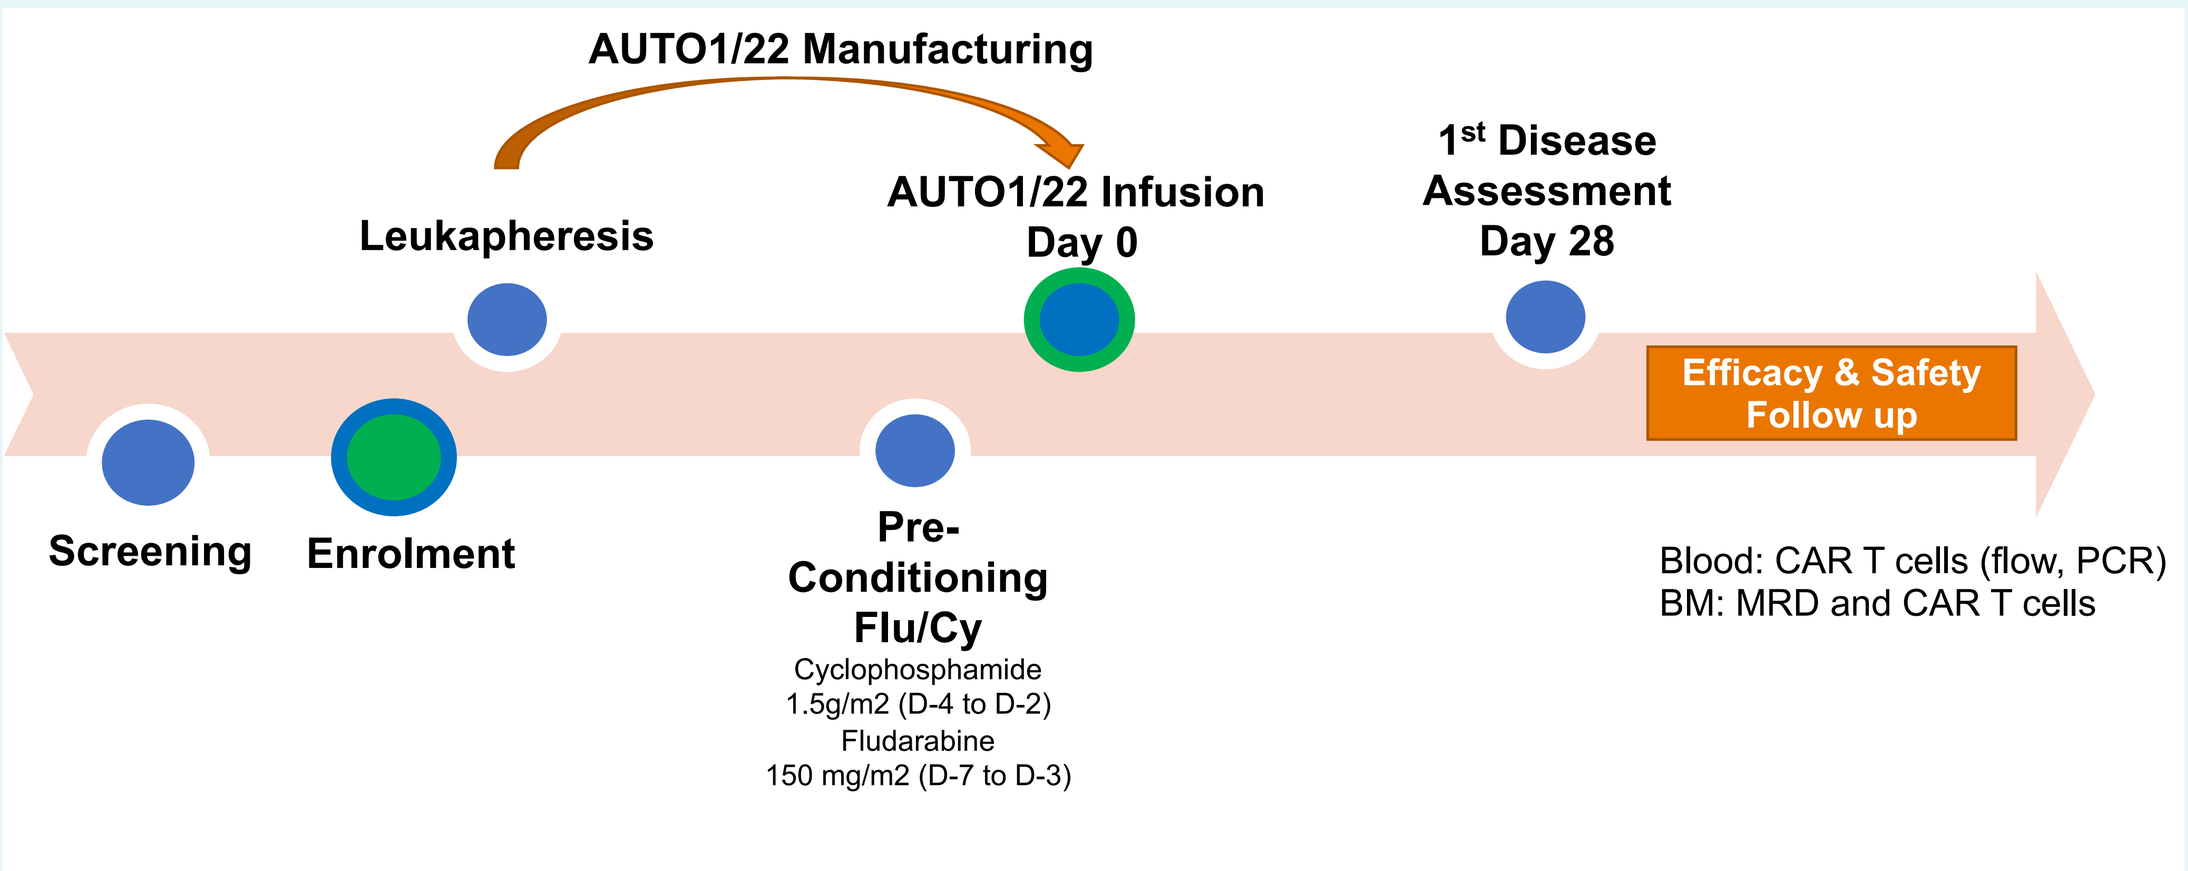
AUTO1/22 Manufacturing
Leukapheresis
AUTO1/22 Infusion
Day 0
1st Disease
Assessment
Day 28
Efficacy & Safety
Follow up
Screening
Enrolment
Pre-
Conditioning
Flu/Cy
Cyclophosphamide
1.5g/m2 (D-4 to D-2)
Fludarabine
150 mg/m2 (D-7 to D-3)
Blood: CAR T cells (flow, PCR)
BM: MRD and CAR T cells

RESULTS

Dual vector transduction of patient leucapheresate gave 3 CAR T cell populations with predominantly central memory phenotype (Tcm) and preservation of stem cell memory T cells (Tscm, Figure 1).

Figure 1

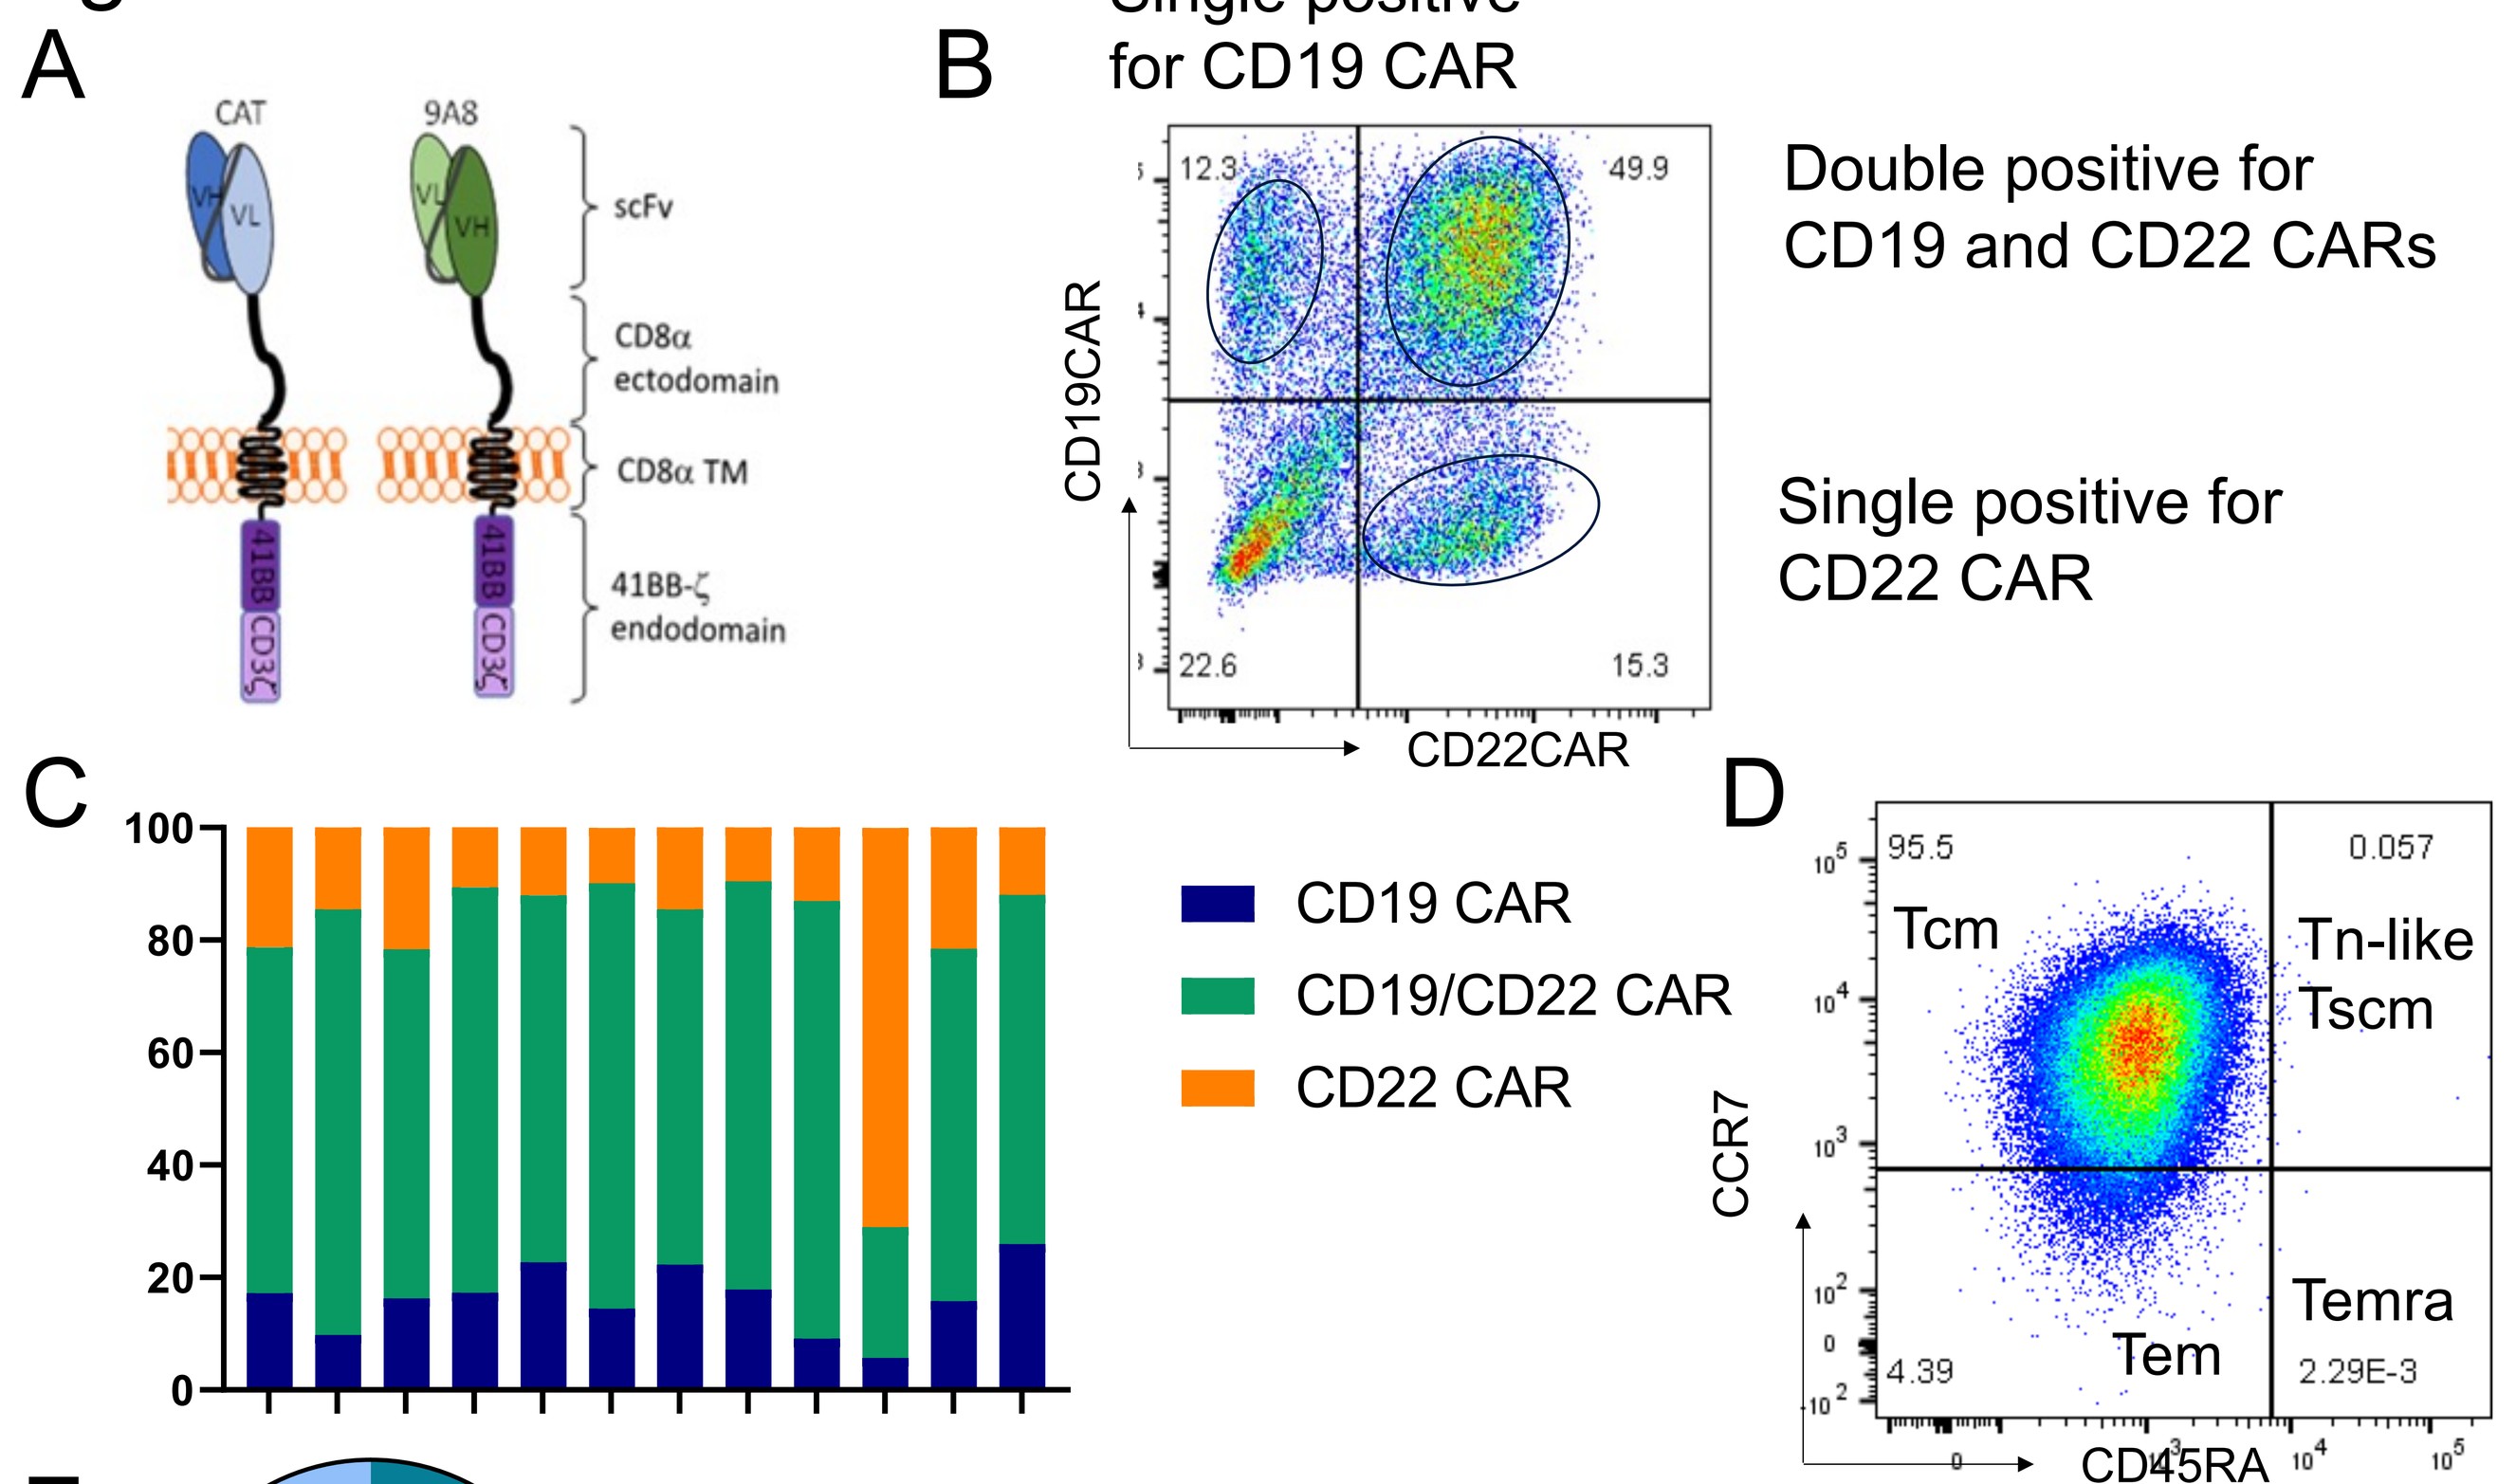
A
CAT
9A8
scFv
CD8α
ectodomain
CD8α TM
41BB-ζ
endodomain
B
Single positive
for CD19 CAR
Double positive for
CD19 and CD22 CARs
Single positive for
CD22 CAR
CD19CAR
CD22CAR
12.3
49.9
22.6
15.3
C
CD19 CAR
CD19/CD22 CAR
CD22 CAR
D
95.5
0.057
Tcm
Tn-like
Tscm
Temra
Tem
4.39
2.29E-3
CCR7
CD45RA

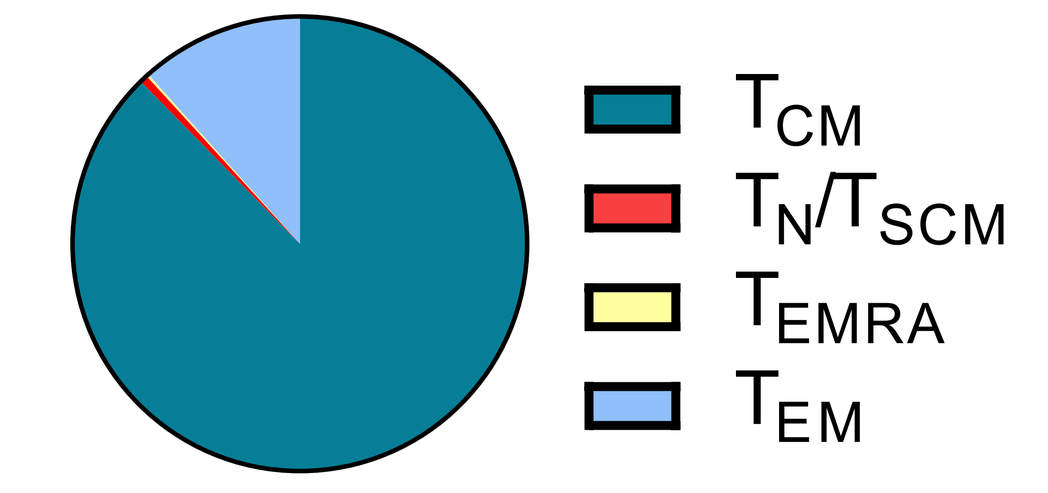
E
TCM
TN/TSCM
TEMRA
TEM

Figure 1. A. CD19 and CD22 CAR architecture. B. Representative CAR expression in patient product. C. Proportions of CAR T cell populations in products. D Representative product memory immunophenotype. E. Summary of median memory subsets of all products

The median transduced cell dose achieved was 664 x 10^6 (range 66-1597), median transduction efficiency was 83% (range 61 - 93), the median VCN was 5.5 (range 3.39-8.00).

Clinical study

13 patients were screened and enrolled (Figure 2), one was withdrawn due to progressive viral infection precluding lymphodepletion. 12 patients were infused (Table 1)

Toxicity

The toxicity profile was favourable (Table 2), tocilizumab was given in 5 pts, there was no severe CRS, no PICU admissions related to CRS. There was 1 grade 4 neurotoxicity - clinically, radiologically and histopathologically-indistinguishable from fludarabine toxicity.

Figure 2 Consort diagram for the cohort

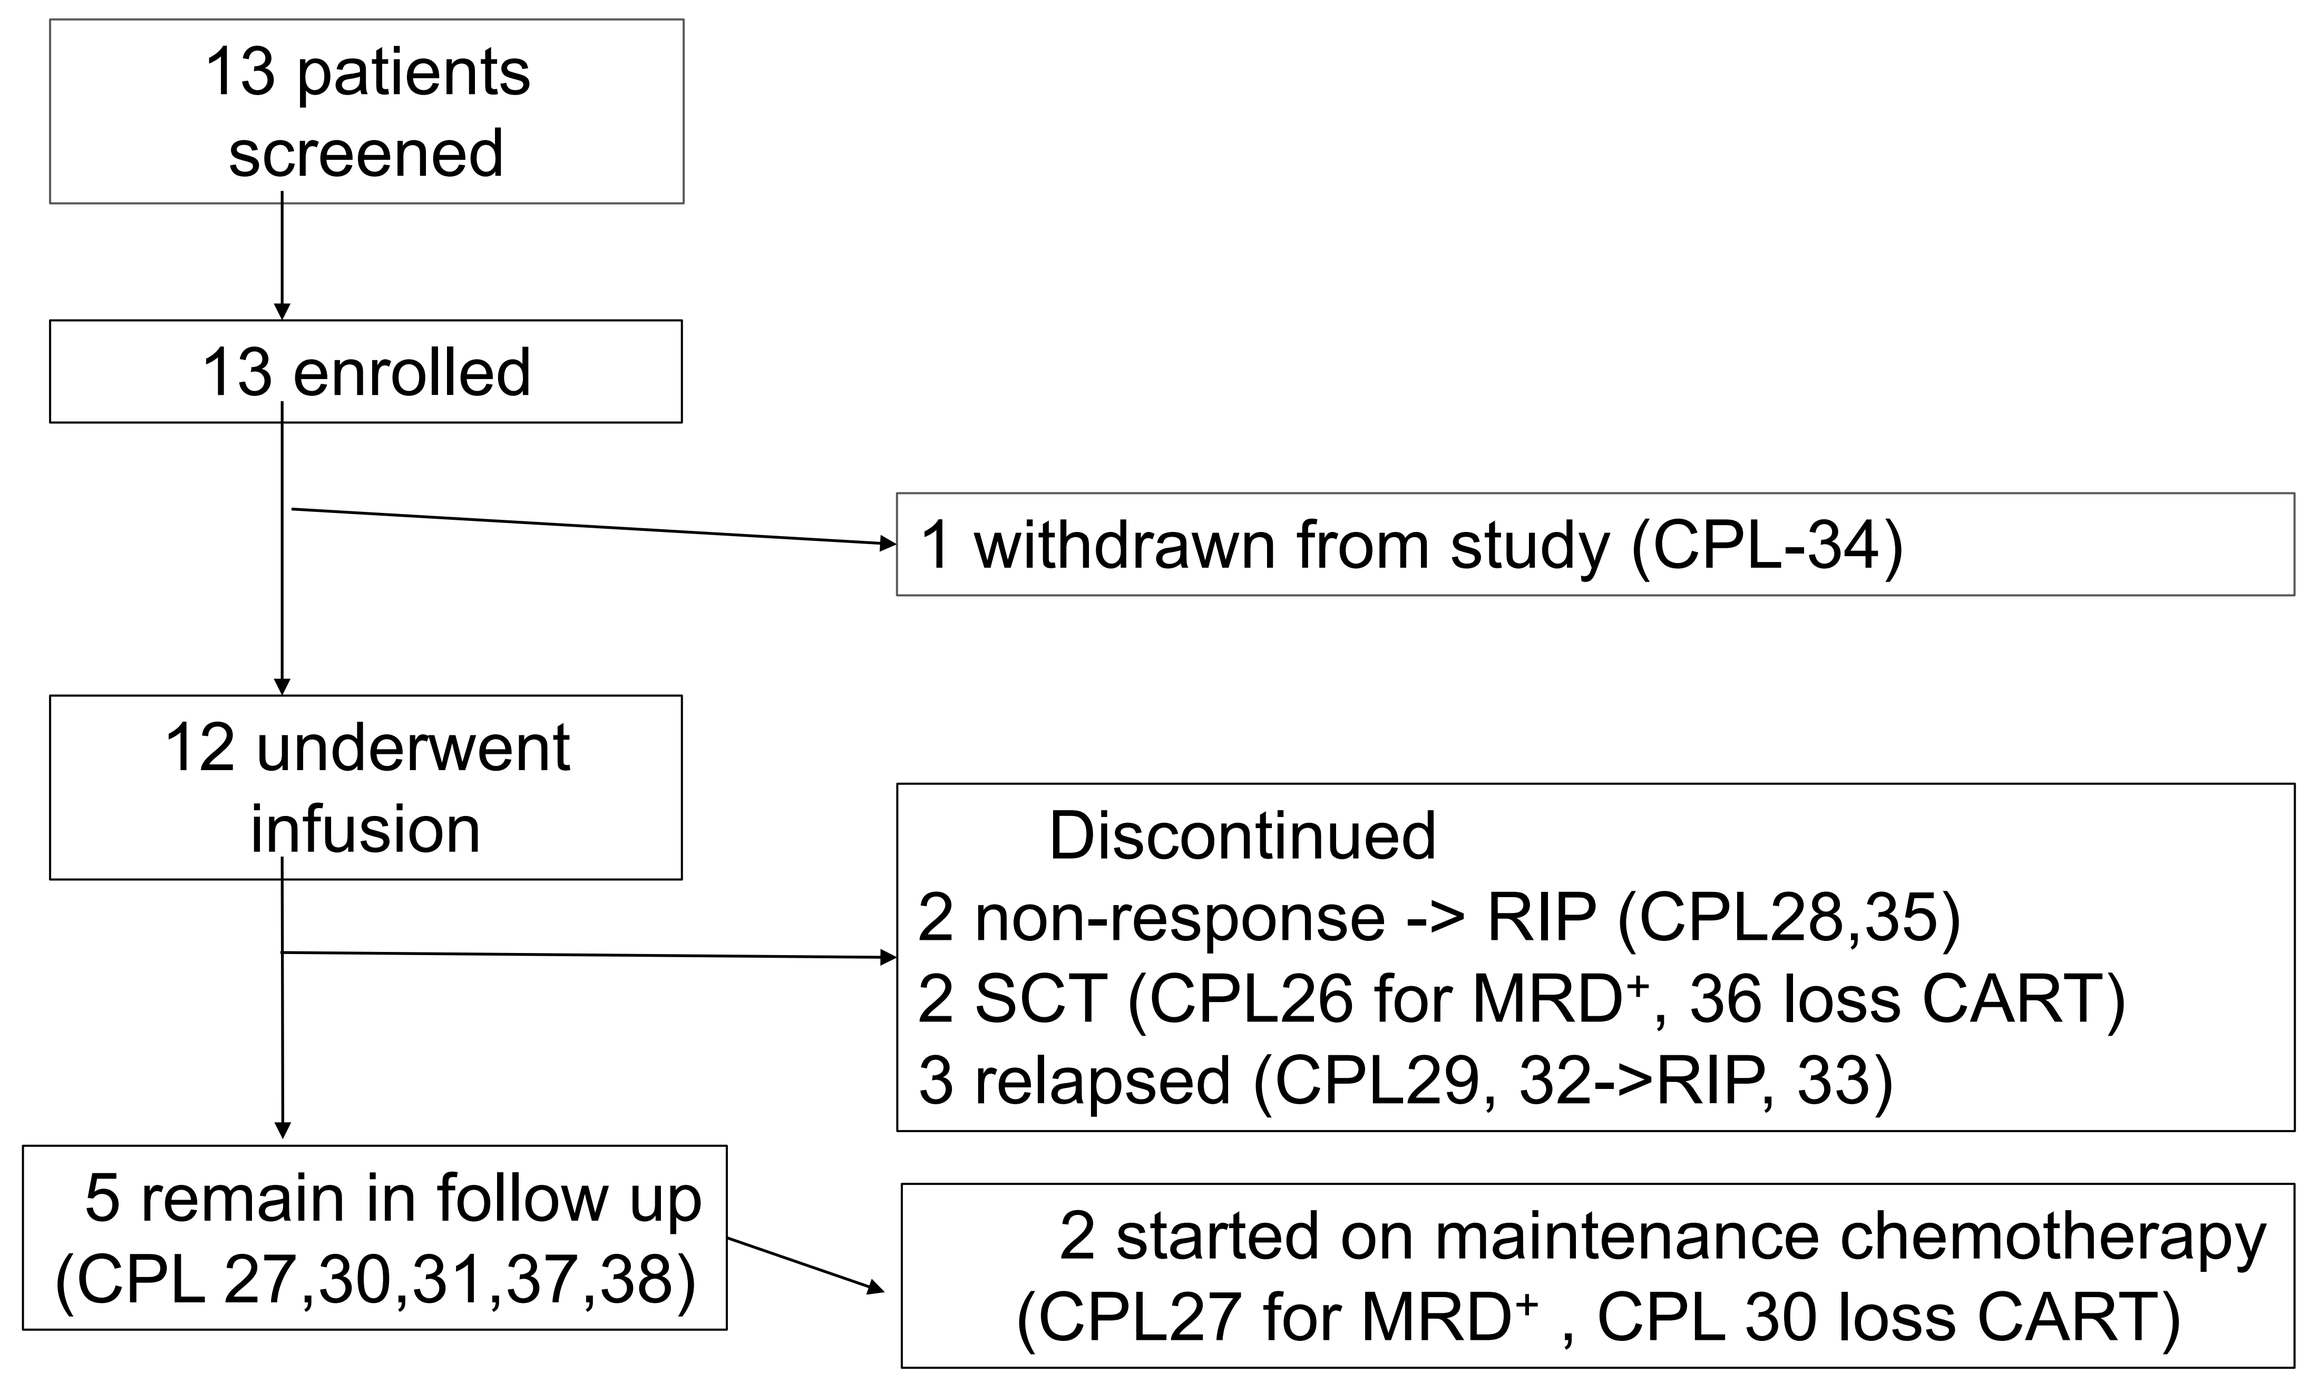
13 patients
screened
13 enrolled
1 withdrawn from study (CPL-34)
12 underwent
infusion
Discontinued
2 non-response -> RIP (CPL28,35)
2 SCT (CPL26 for MRD+, 36 loss CART)
3 relapsed (CPL29, 32->RIP, 33)
5 remain in follow up
(CPL 27,30,31,37,38)
2 started on maintenance chemotherapy
(CPL27 for MRD+ , CPL 30 loss CART)

Table 1 Patient characteristics

Total	n=12
Median age at registration	12 yrs (range = 3.7-20.5)
Indication	
Post SCT relapse	**6 (55%, 3 isolated extramedullary)**
1st relapse	2 (18%)
2^{nd} relapse	8 (73%)
>2^{nd} relapse	1 (9%)
Median number of lines of prior Rx	**3 (range 2-6)**
Prior Inotuzumab/Blinatumomab	6 (55%)
Prior CD19 CAR T cell therapy	**4 (36%)**
Prior CD19-ve disease	**3 (27%)**
BM status pre-lymphodepletion	
Morphological relapse (>5% blasts)	4 (36% + 1 NE with 6% mol MRD)
MRD 2-5%	1 (9%)
MRD 10^{-2}-10^{-5}	3 (27%)
MRD negative	3 (18%)

Table 2 Toxicity

Criteria	N=12 N (%)
CRS Maximum Grade (ASTCT)	
Grade 1	5 (41%)
Grade 2	6 (50%)
Grade 3-4	0
ICANS Maximum Grade (ASTCT)	
Grade 1	4 (36%)
Grade 2	1 (8%)
Grade 3	0
Grade 4 (MRI leukoencephalopathy)	1 (8%)
Cytopenia not resolving by or recurring after day 28 Maximum Grade (CTCAE)	
Grade 1-3	3 (25%)
Grade 4	8 (67%)

Criteria:	N=12 N (%)
B cell aplasia	
At day 30	11 (92%)
At last follow up	7 (58%)

Cytopenias were prominent: 10/12 grade 3-4 cytopaenias persisting beyond / recurring after 30 days. In 8/10 cases, this had resolved by the data cut-off**,** 1 patient had a $CD34^+$ top up.

Disease response and outcomes

10 out of 12 MRD- CR/CRi/CCR, 2 NR (including 1 patient with CD19 negative disease and Li Fraumeni syndrome, Table 3). There have been 3 relapses and emergence of MRD in 3 patients (all CD19/CD22 positive). Overall and event free survival are shown in Table 4 and Figure 3

Table 3 Disease response

Total	N=11
Molecular MRD neg CR/Cri/CCR by d30	10 (83%)
Disease progression	2 (17%)
Relapse	
Ag negative relapse	0
CD19+/CD22+ relapse	3
Emergence of MRD	2

Figure 3 Patient outcomes

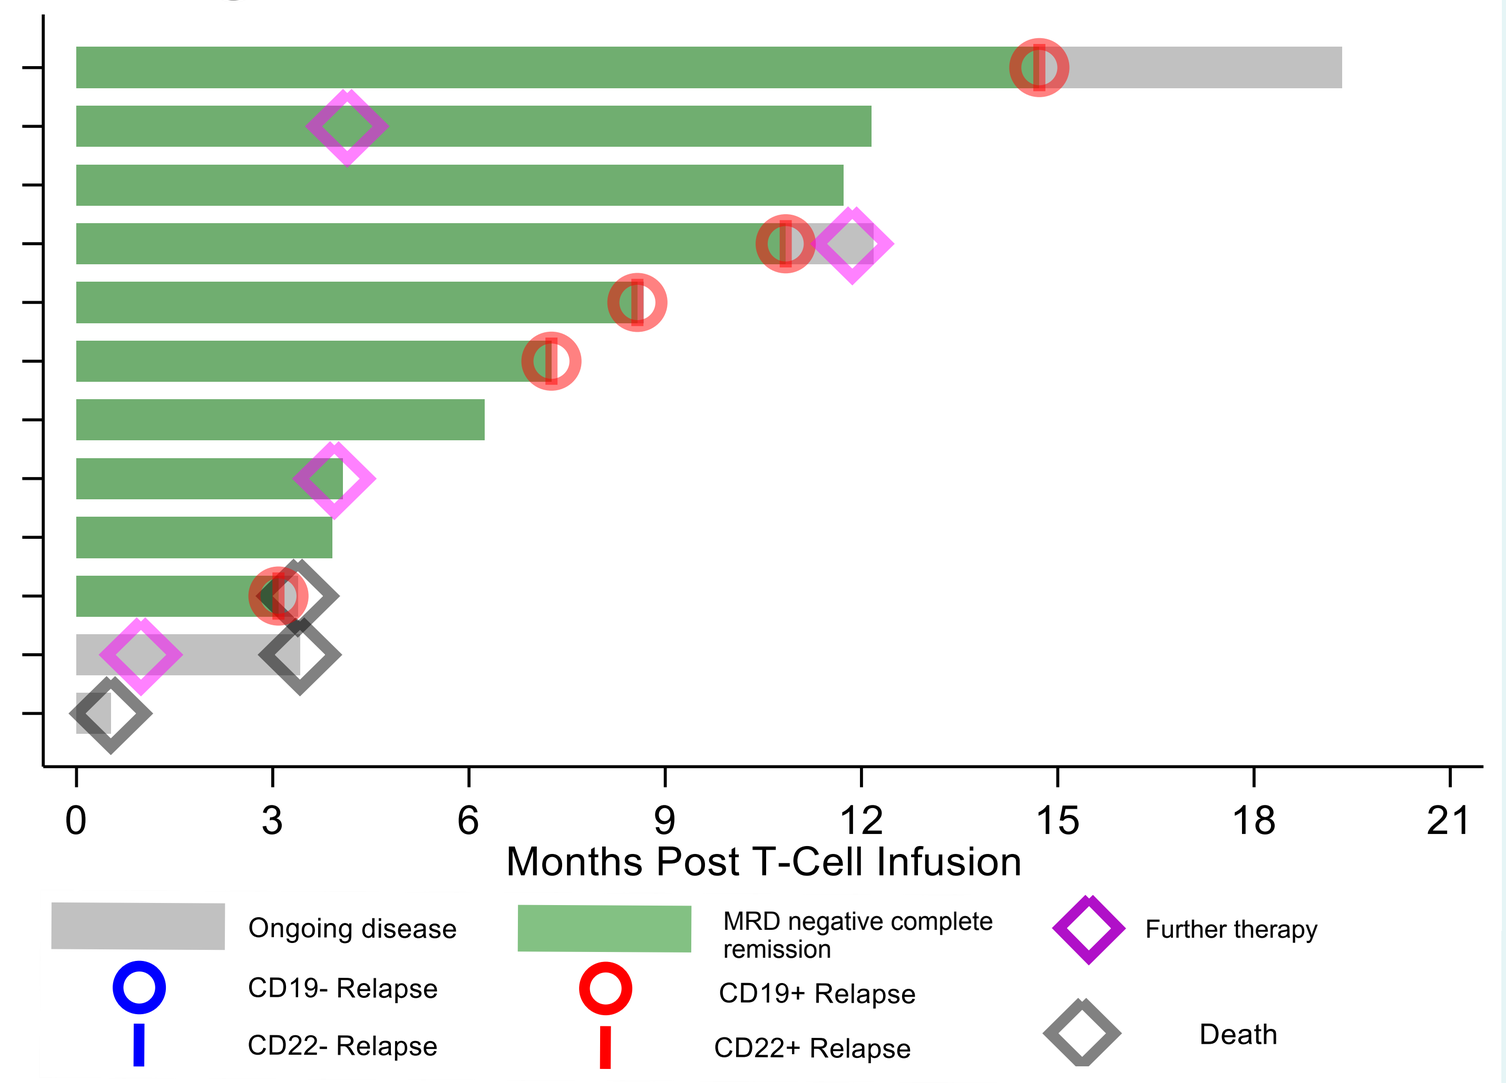
0
3
6
9
12
15
18
21
Months Post T-Cell Infusion
Ongoing disease
MRD negative complete remission
Further therapy
CD19- Relapse
CD22- Relapse
CD19+ Relapse
CD22+ Relapse
Death

Table 4 Outcomes

	Overall Survival	Event-Free Survival	"Stringent" Event-Free Survival
	N=12	**N=12**	**N=12**
N. of events	3 (25%)	4 (33%)	7 (58%)
12 month (95%CI)	**75%** (41% - 91%)	**60%** (23% - 84%)	**38%** (9% - 67%)

Figure 4 Survival curves

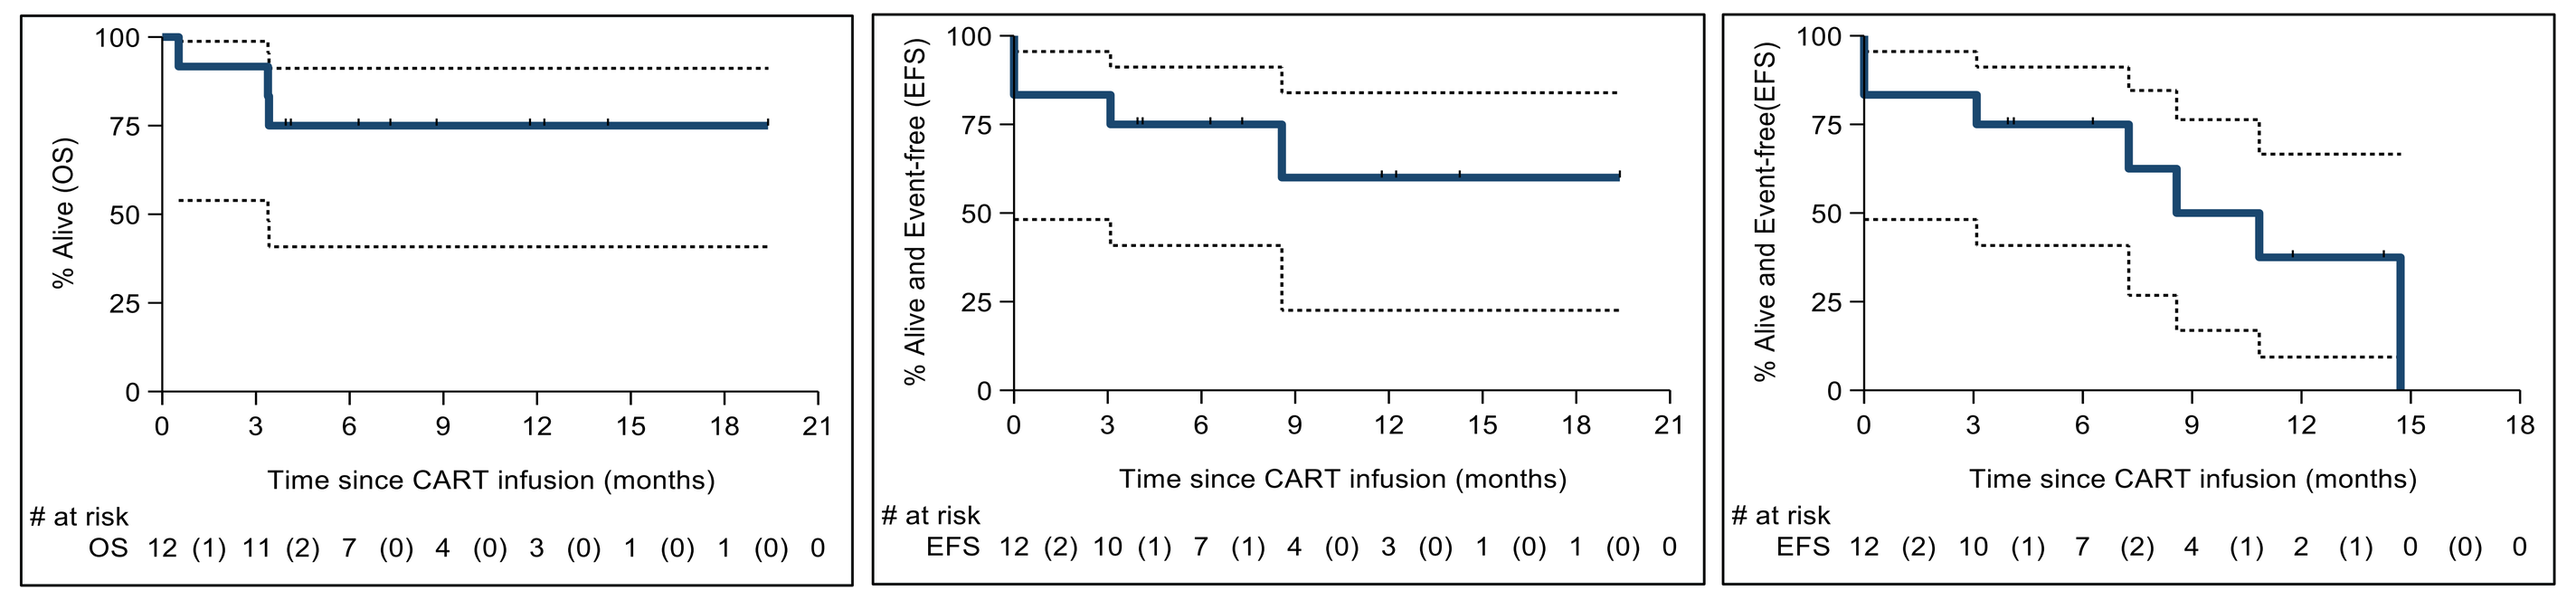
% Alive (OS)
Time since CART infusion (months)
at risk
OS 12 (1) 11 (2) 7 (0) 4 (0) 3 (0) 1 (0) 1 (0) 0
% Alive and Event-free (EFS)
Time since CART infusion (months)
at risk
EFS 12 (2) 10 (1) 7 (1) 4 (0) 3 (0) 1 (0) 1 (0) 0
% Alive and Event-free(EFS)
Time since CART infusion (months)
at risk
EFS 12 (2) 10 (1) 7 (2) 4 (1) 2 (1) 0 (0) 0

Overall Survival | **Event-free survival (ELIANA)** | **Stringent Event-free survival**

Conclusions

Safety profile was favourable: no severe CRS

Excellent CART expansion, including of CD22 CAR population

10/12 MRD negative CR/CRi/CCR, **despite poor risk cohort (4 Kymriah failures, 3 CD19neg disease, 3 non-CNS EM disease)**

1 yr EFS 60% **equivalent** to ELIANA

Median **7.5mo duration** of persistence of CD22 CAR

No antigen-ve relapse seen in responding patients

At median FU 8.7 months, 5/10 responding patients in MRD-ve CR (4-12 mo), 2 after further therapy for early loss CAR T persistence

s.ghorashian@ucl.ac.uk

NIHR | Great Ormond Street Hospital Biomedical Research Centre
NIHR | University College London Hospitals Biomedical Research Centre
NHS Great Ormond Street Hospital for Children NHS Foundation Trust